DENKI (Dual ENKephalinase Inhibitors), a new class of painkillers devoid of abuse potential and of opiate side-effects

Hervé Poras[a], Gwendolyn E Burgess[b], Bryan F Sears[b], Tanja Ouimet[a], Michel Wurm[a], Marie-Claude Fournié-Zaluski[a], Bernard P Roques[a], Emily Jutkiewicz[b]

[a] Pharmaleads, Paris BioPark, 11 rue Watt, 75013 Paris, FRANCE; [b] Department of Pharmacology, University of Michigan, Medical School, Ann Arbor, USA

Introduction: DENKI's mechanism of action

Following the opioid crisis, painkillers with novel mechanisms of action are being sought as alternatives to opioids and non-opioids / NSAIDS. Innovative non-opiate and non-addictive products are developed for the management of acute and chronic severe pain, a growing market with significant unmet medical need. Pharmaleads develops **new efficacious analgesics acting selectively when and where enkephalins are released after painful stimulus**, **enhancing the power of endogenous opioid peptides, a key element of the body's natural pain modulation system. Dual ENKephalinase inhibitors (DENKI®) protect enkephalins from their rapid enzymatic degradation, hence increasing their local concentrations. The resulting physiological analgesia provides a safe and highly effective new pain management option.**

Enkephalins are the most abundant endogenous opiates; they are widely expressed and bind both to delta and mu opioid receptors. They induce analgesia centrally and peripherally by inhibiting pain signaling.

Peripherally, they suppress tetrodoxin-resistant Na^+, TRPV1 and other cation currents stimulated by inflammatory agents. Through these effects, they attenuate peripheral nociceptor terminal excitability, the propagation of action potentials and the release of excitatory pro-inflammatory neuropeptides such as SP and CGRP.

Once released, the opioid peptides Met-enkephalin, YGGFM, and Leu-enkephalin, YGGFL, are inactivated by two zinc-metallopeptidases, neutral endopeptidase or neprilysin (NEP, EC 3.4.24.11) and aminopeptidase N (APN, EC 3.4.11.2), providing the opportunity to design enzyme inhibitors as **new efficacious analgesics acting selectively when and where enkephalins are released after noxious stimulus.**

However, inhibiting only one of these enzymes does not increase enkephalin concentrations sufficiently to induce analgesic response[1]. We have therefore developed the concept of **"dual inhibitors"** which **inhibit both NEP and APN[1].** Accordingly, several families of dual inhibitors were designed and synthetized[1-3] and shown to be very active by various administration routes.

Two Pharmaleads' Dual ENKephalinase Inhibitors (DENKI®), **PL37** and **PL265**, are now in clinical development as new analgesics devoid of the side effects of morphine and derivatives. Both are active *per os* without entering the CNS while i.v. PL37 crosses the blood brain barrier, when properly formulated. Their efficacy was assessed in a wide array of pain models and their safety / tolerability were evaluated, in particular with regards to opioid-like side-effects.

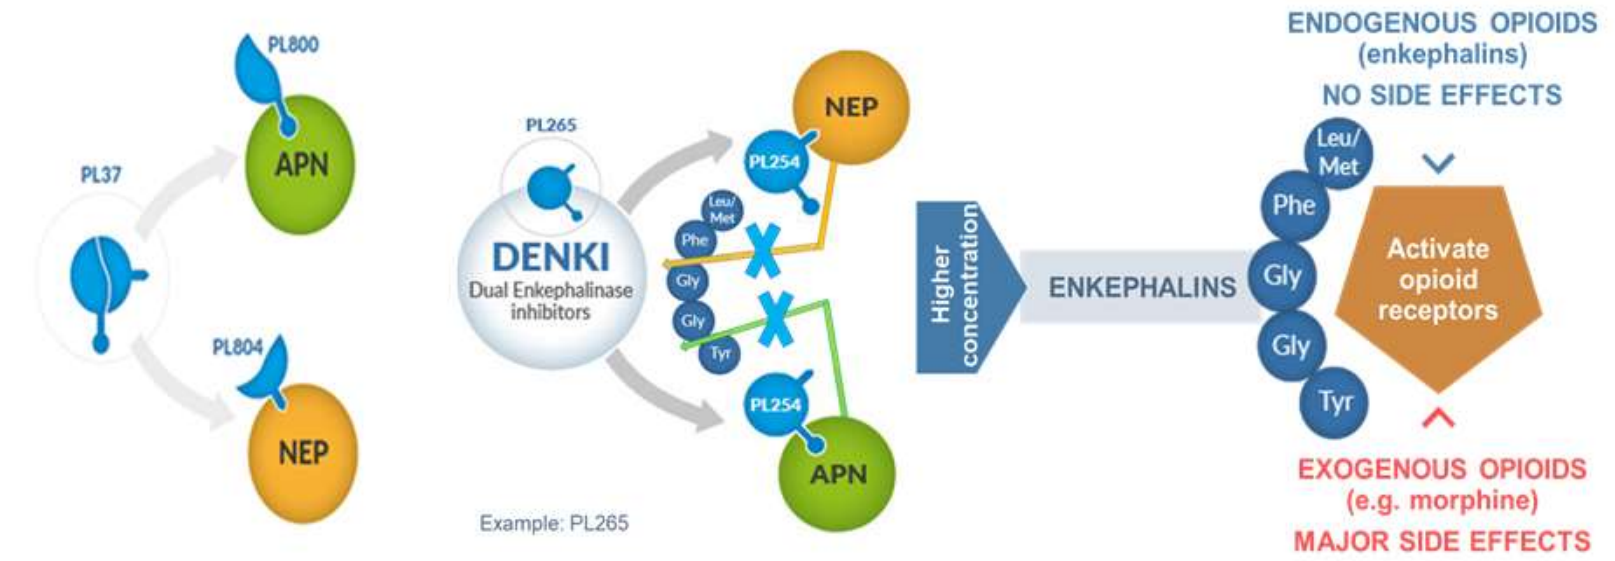
PL800
APN
PL37
PL804
NEP
PL265
DENKI
Dual Enkephalinase inhibitors
PL254
NEP
Leu/Met
Phe
Gly
Gly
Tyr
PL254
APN
Example: PL265
Higher concentration
ENKEPHALINS
ENDOGENOUS OPIOIDS
(enkephalins)
NO SIDE EFFECTS
Leu/Met
Phe
Gly
Gly
Tyr
Activate opioid receptors
EXOGENOUS OPIOIDS
(e.g. morphine)
MAJOR SIDE EFFECTS

DENKI® protect enkephalins from degradation

We previously have shown that DENKI® (P8B), ip injected in an inflammatory model of pain, **dose-dependently increase enkephalin concentrations**[4] due to inhibition of the degrading enzymes APN and NEP, inducing analgesic response. Experiments were performed 4 days after triggering inflammation.

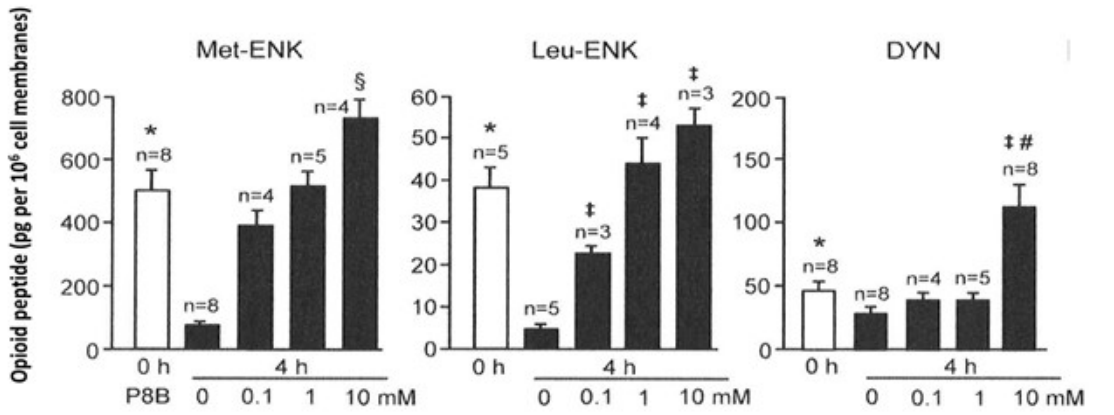
Opioid peptide (pg per 10⁶ cell membranes)
Met-ENK
Leu-ENK
DYN
0 h
4 h
P8B 0 0.1 1 10 mM
n=8
n=4
n=5

Sciatic nerve suspensions from the rats were incubated for 15 min with inhibitors before addition of opioid peptides (0.4 –22.3 nM). Opioid concentrations were measured immediately thereafter (0 h; open bars) and 4 h following coincubation with nerves (filled bars at different concentrations of P8B).

Radioimmunoassays have shown that *ex vivo* inhibition of APN and NEP, by an aminophosphinic DENKI® (P8B), in leukocytes as well as these present on peripheral nerves from inflamed paws, prevents the degradation of enkephalins and dynorphin, secreted in rats' hindpaw after intraplantar administration of complete Freund's adjuvant (150 µL).

These results were well correlated with the analgesic response.

Thus, leukocytes and peripheral nerves are important sources of APN and NEP in inflamed tissue, and their blockade promotes peripheral opioid analgesia.

Methods

Animal experiments are carried out in accordance with the European Communities Council directive (89/609/ECC) and with the ethical guidelines of the International Association for the Study of Pain (IASP).

Drug Discrimination: Rats are trained to discriminate either 3 mg/kg morphine (mu agonist) or 3 mg/kg SNC80 (delta agonist) from saline under a FR10 schedule for sucrose delivery. All injections are administered prior to entering the operant chamber during a 20-min blackout period. Rats can respond for as many sucrose pellets as possible in a 10-min period of responding. On training days, responding on the injection-appropriate nose poke is reinforced. On test days, responding on either nose poke results in delivery of sucrose pellets. After meeting discrimination criteria, single doses of various drugs are substituted. Rats are tested at most twice per week.

Self-administration: Rats are trained to self-administer 0.003 mg/kg/infusion remifentanil on a FR1 schedule of reinforcement during 60 min daily sessions. After approximately 7 days, morphine is substituted at a dose of 0.3 mg/kg/infusion during 180 min daily sessions. Once stable responding is achieved, saline is repeatedly and intermittently substituted until rats decrease responding to <20% of that observed in the presence of morphine. Then different doses of morphine, PL37, or PL265 are substituted for at least 3 consecutive days.

Forced swim test: Rats are injected with PL37 or PL265 i.p. 30 min prior to the forced swim test. Counts of immobility, swimming, and climbing are later scored by an individual blind to the experimental conditions.

Safety & tolerability with respect to opiate-like side effects

Despite their undisputable efficacy in acute pain, management using morphine and its derivatives elicits dose limiting unwanted side effects (nausea, constipation, sedation, respiratory depression) while their long term use carries with it an elevated risk for increasing tolerance and addiction.

The following experiments were performed in animals to study these opiate abuse potential side effects in case of use of DENKI® as a new pharmacological class of painkillers.

Self-administration paradigm in rats

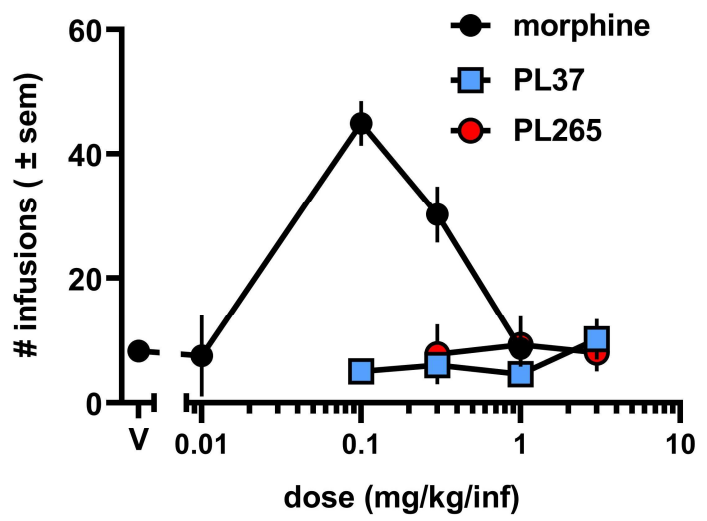
morphine
PL37
PL265
infusions (± sem)
60
40
20
0
V 0.01 0.1 1 10
dose (mg/kg/inf)

Morphine produces an inverted U-shaped dose effect curve as expected.
PL37 fails to maintain responding, producing levels similar to saline.
PL265 maintains responding in 2 rats thus far, but at a dose of 1 mg/kg/infusion.
While PL265 maintains responding, the levels observed are significantly lower than those maintained by morphine.

No discriminative effect in rats

In rats trained to discriminate morphine (3 mg/kg), the MOR agonist hydrocodone generalizes to the discriminative stimulus effects of morphine.
PL37 and PL265 do not generalize to morphine, suggesting that these compounds do NOT have interoceptive effects (e.g subjective effects) similar to morphine.

In rats trained to discriminate SNC80 (3 mg/kg), the SNC80 analog AZD2327 fully generalizes to the discriminative stimulus effects of SNC80, suggesting similar interoceptive effects (note: these 2 compounds produce convulsions).
PL37 and PL265 produce partial generalization.
Neither compound produces convulsions nor decreases response rates consistently in all rats.

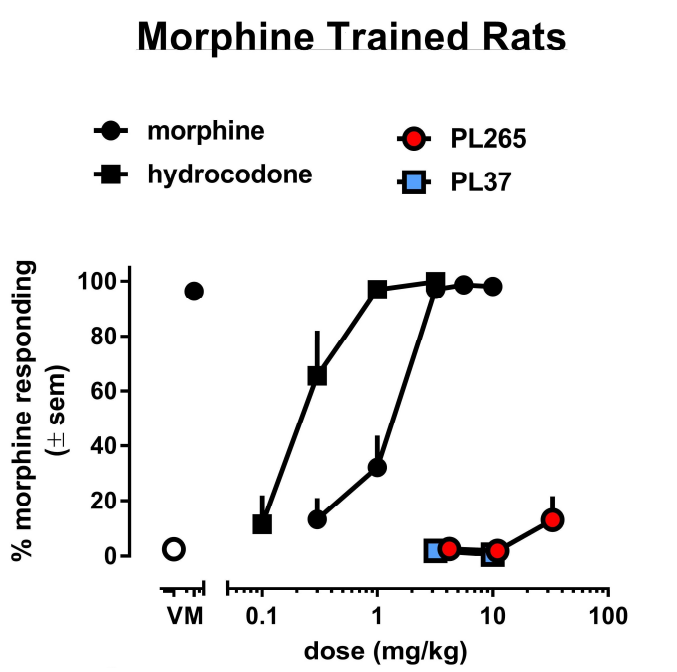
Morphine Trained Rats
morphine
hydrocodone
PL265
PL37
% morphine responding (± sem)
100
80
60
40
20
0
VM 0.1 1 10 100
dose (mg/kg)

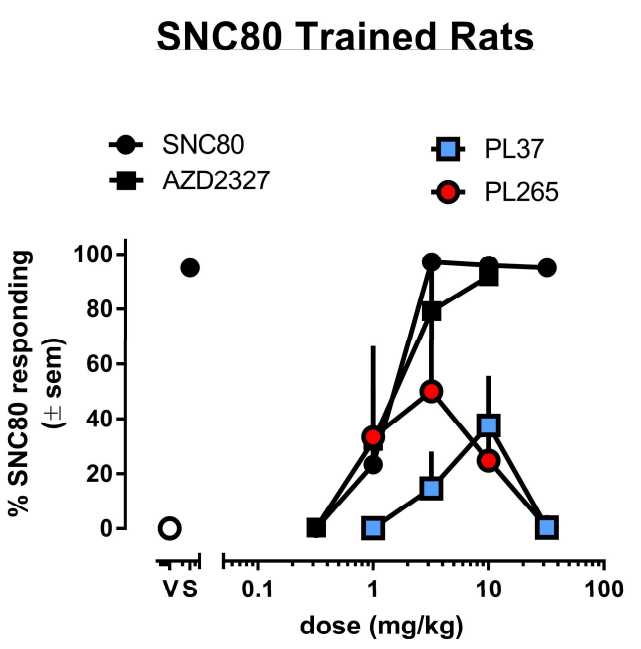
SNC80 Trained Rats
SNC80
AZD2327
PL37
PL265
% SNC80 responding (± sem)
100
80
60
40
20
0
VS 0.1 1 10 100
dose (mg/kg)

Forced Swim Test – Antidepressant-like activity

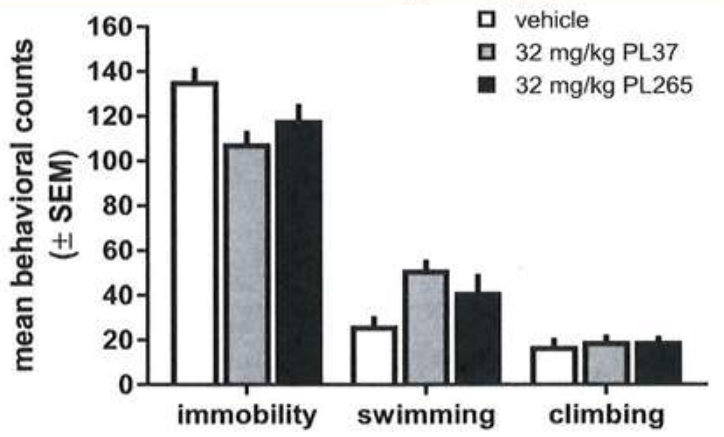
vehicle
32 mg/kg PL37
32 mg/kg PL265
mean behavioral counts (± SEM)
160
140
120
100
80
60
40
20
0
immobility
swimming
climbing

Opioid peptides have been demonstrated to have antidepressant actions in animal models of depression without producing convulsions. For example, **enkephalins decreased immobility in the forced swim test and in the learned helplessness paradigm in rats, demonstrating antidepressant-like effects.[5]**

The DENKI® RB101 appears to produce antidepressant-like effecs similar to those observed with nonpeptidic delta-opioid receptor agonists.[6]

PL37 and PL265 decrease immobility in the rat forced swim test; however, this effect was only significant with PL37.

Conclusions

Across a wide array of animal models of pain, DENKI® induce antinociceptive responses similar to morphine. DENKI® remain active after chronic administration of morphine, indicating that DENKI®-induced analgesia persists in morphine-tolerant mice. Moreover, DENKI® do not induce constipation in animals or humans. Finally, **PL37 and PL265 are both shown as devoid of any abuse liability even at high doses after repeated administration and could be used as a new class of painkillers in all types of severe pain and as an affordable solution to the opioid crisis.**

Based on these remarkable pharmacological results, DENKI® are currently in clinical development as novel painkillers. PL265 is being developed for the oral treatment of neuropathic pain, osteoarthritis pain or topical treatment of ocular / inflammatory pain whereas PL37 targets the treatment of post-surgical / traumatic / breakthrough cancer pain as a substitute to injectable opiates.

References: [1] Roques B.P., Fournie-Zaluski MC., Wurm M., Nature Rev. Drug Discov., 2012, 11, 292-310; [2] Poras H., Bonnard E., Dangé E., Fournie-Zaluski MC., Roques B.P., J. Med. Chem., 2014, *57*, 5748-5763; [3] Bonnard E., Poras H., Nadal X., Maldonado R., Fournié-Zaluski M.C., Roques B.P., Pharmacol. Res. Perspect., 2015, *3,* e00116, doi: 10.1002/prp2.116; [4] Schreiter A., Core C,., Labuz D., Fournie-Zaluski MC., Roques B.P., Stein C., Machelska H., FASEB J., 2012, 26, 5161-5171; [5] Kastin A.J., Scollan E.L., Ehrensing R.H., Schally A.V., Coy D.H., Pharmacol. Biochem., 1978, 9, 515-519; [6] Jutkiewicz E;M., Torregrossa M.M., Sobczyk-Kojiro K., Mosberg H.I., Folk J.E., Rice K.C., Watson S.J., Woods J.H., Eur. J. Pharmacol, 2006, 531, 151-159.