DENKI (Dual ENKephalinase Inhibitors), a new class of painkillers devoid of abuse potential and of opiate side-effects

Michel Wurm[1], Tanja Ouimet[1], Nicolas Marie[2], Florence Noble[2], Rafael Maldonado[3], Emily Jutkiewicz[4], Hervé Poras[1]

[1] Pharmaleads SA, Paris, France; [2] CNRS ERL 3649, Inserm UMR-S 1124, Centre Universitaire des Saints Pères, Paris, France;

[3] Laboratori de Neurofarmacologia, Universitat Pompeu Fabra, Barcelona, Spain; [4] Department of Pharmacology, University of Michigan, Medical School, Ann Arbor, USA

Introduction: DENKI's mechanism of action

Opioid peptides are widely distributed in numerous organs and tissues. Their presence has been detected both in the central nervous system and in peripheral structures that regulate fundamental functions of the body.

Recently, **human congenital insensitivity to pain** due to loss of function of $Na_v1.7$ has been **directly linked to a sustained increase in enkephalin expression.**[1]

The opioid peptides Met-enkephalin, YGGFM, and Leu-enkephalin, YGGFL, are inactivated by two zinc-metallopeptidases, NEP (EC 3.4.24.11)[2] and aminopeptidase N, APN (EC 3.4.11.2)[1] providing the opportunity to design enzyme inhibitors as **new efficacious analgesics** acting selectively when and where enkephalins are released after noxious stimulus.

However, inhibiting only one of these enzymes does not increase enkephalins concentrations enough to induce **analgesic responses.**[2] We have therefore developed the concept of "dual inhibitors" which inactivate **both NEP and APN.**[2] Accordingly, several families of dual inhibitors have been synthetized[2, 3, 4] and were shown to be very active by various administration routes.

Two Pharmaleads' Dual ENKephalinase Inhibitors (DENKI) are now in clinical development as new analgesics devoid of the side effects of morphine and derivatives. Both are active *per os* without entering the CNS while i.v. PL37 crosses the blood brain when properly formulated. Their efficacy was assessed in a wide array of pain models and their safety/tolerability were evaluated, generally and with regards to their opioid side effects.

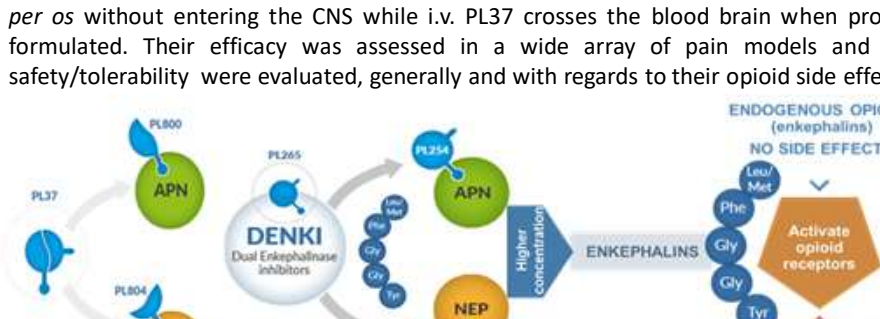
PL37
PL800
APN
PL804
NEP
PL265
DENKI
Dual Enkephalinase inhibitors
PL254
APN
NEP
Higher concentration
ENKEPHALINS
Leu/Met
Phe
Gly
Gly
Tyr
ENDOGENOUS OPIOIDS (enkephalins)
NO SIDE EFFECTS
Activate opioid receptors
EXOGENOUS OPIOIDS (e.g. morphine)
MAJOR SIDE EFFECTS
Example: PL265

We previously showed that DENKI injected in an inflammatory model of pain showed a dose dependent **increase in enkephalin concentrations**[5] due to inhibition of the degrading enzymes APN and NEP.

A single dose of aminophosphinic dose-dependently prevents the degradation of ENK on peripheral nerves from inflamed paws

Radioimmunoassays showing that inhibition of leukocytic APN and NEP by an aminophosphinic DENKI prevents the degradation of enkephalins, secreted in rats' hindpaw after intraplantar administration of complete Freund's adjuvant (150µL). Experiments were performed 4 days after triggering inflammation.[5]

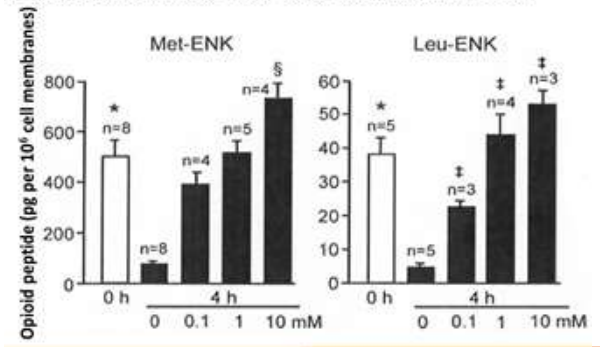
Met-ENK
Leu-ENK
Opioid peptide (pg per 10^6 cell membranes)
0 h
4 h
0 0.1 1 10 mM

APN inhibition in human plasma (single dose) | NEP inhibition in human plasma (single dose)

Oral administration of **PL265 in human (Ph1 Single Ascending Dose)** inhibits APN and NEP activity in plasma.[6]

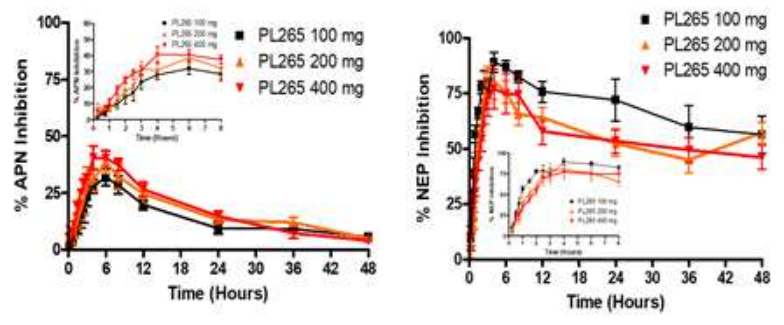
PL265 100 mg
PL265 200 mg
PL265 400 mg
% APN Inhibition
% NEP Inhibition
Time (Hours)

Methods

Experiments were performed in male OF1 mice, 20−32 g, and male Sprague−Dawley rats, 200−280 g. Animals were housed for at least 2 days before experiments in a room with controlled temperature (21 ± 2 ° C) and a 12 h alternating light−dark cycle. Food and water were available *ad libitum*. Great care was taken to avoid or minimize discomfort of animals. Animal experiments were carried out with the European Communities Council directive (89/609/CEE) and in accordance with the ethical guidelines of the IASP. The maximal volume administered was 0.1 mL/100g for intravenous injection and 0.2 mL/100g. orally.

Animal pharmacology: efficacy on neuropathic, inflammatory and nociceptive pain

PL37 i.v. is active in the hot plate test, the tail-flick test (acute pain),[3] the acetic acid-induced writhing model (chronic pain), a cancer pain model and the Brennan model (post-operative pain model).[7]

PL265 is orally active on formalin and complete Freund's adjuvant (CFA) models (inflammatory pain), chronic constrictive injury (CCI), partial sciatic nerve ligation (PSNL) (neuropathic pain) [4] and ocular pain/inflammation models.

Additionally, DENKIs do not elicit any of the side-effects of opiates such as tolerance and addiction or constipation.

Oral PL265: neuropathic pain model – PSNL in mice

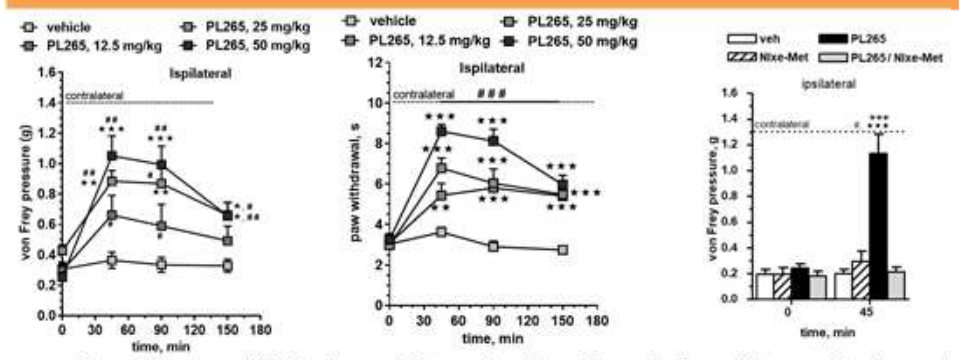
vehicle
PL265, 12.5 mg/kg
PL265, 25 mg/kg
PL265, 50 mg/kg
Ispilateral
contralateral
von Frey pressure (g)
paw withdrawal, s
time, min
veh
PL265
Nlxe-Met
PL265 / Nlxe-Met
ipsilateral

Dose-dependent antiallodynic and hyperalgesic effects induced by a single oral administration of PL265 in mice lasted at least 150 min. The antiallodynic effect at PL265 50 mg/kg wax fully reversed by co-injection of naloxone methiodide (5 mg/kg, i.p.), indicating that only peripheral opioid receptors are recruited (right histogram).

Oral PL265: inflammatory pain model (formalin) in mice

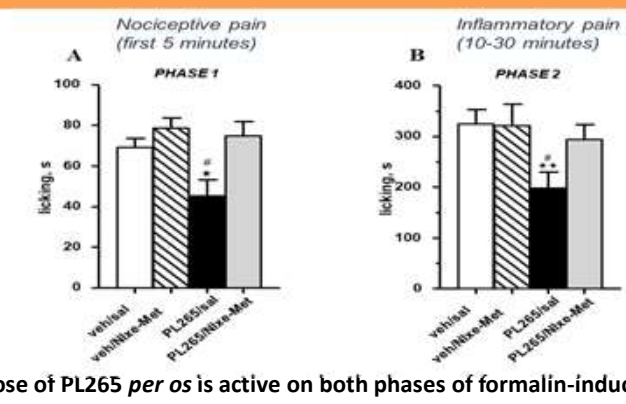
Nociceptive pain (first 5 minutes)
Inflammatory pain (10-30 minutes)
PHASE 1
PHASE 2
licking, s
veh/sal
veh/Nlxe-Met
PL265/sal
PL265/Nlxe-Met

A single dose of PL265 *per os* is active on both phases of formalin-induced pain.

iv PL37: acute central pain – HPT in mice

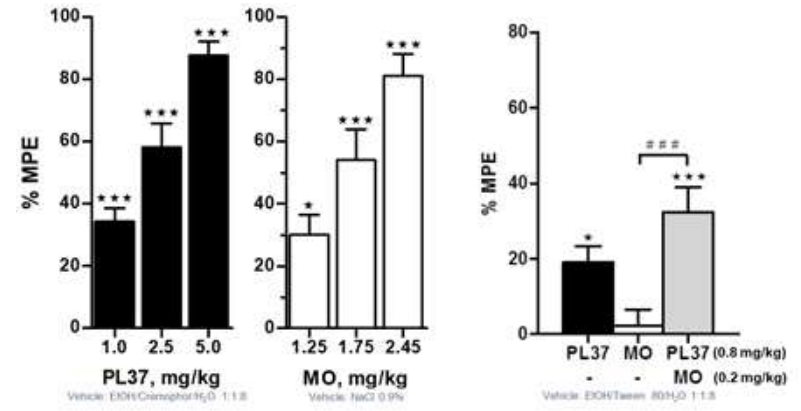
% MPE
PL37, mg/kg
MO, mg/kg
PL37 MO PL37 (0.8 mg/kg)
MO (0.2 mg/kg)

Both compounds, PL37 and morphine (MO), induced dose-dependent high antinociceptive responses with close ED_{50}s of 1.8 (1.2-2.6) and 1.6 (1.4-2.0) mg/kg for PL37 in EtOH/cremophor/water and MO in saline respectively (left side).

A significant increase in antinociceptive response was observed by co-administration of PL37 (0.8 mg/kg) and MO (0.2 mg/kg) by i.v. route (right side), suggesting a synergistic effect.

iv PL37: visceral pain - acetic acid-induced writhing test in rats

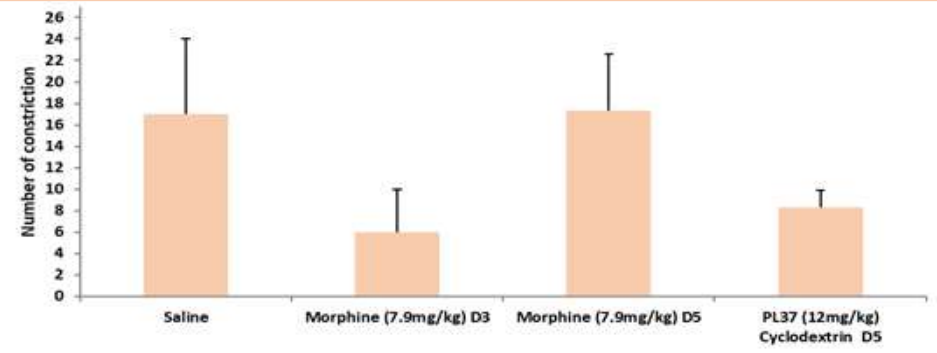
Number of constriction
Saline
Morphine (7.9mg/kg) D3
Morphine (7.9mg/kg) D5
PL37 (12mg/kg) Cyclodextrin D5

A 5-day continuous intravenous infusion of PL37 protects against acetic acid-induced abdominal constrictions in the rat. Tolerance to morphine appears between D3 and D5 while PL37 remains active at D5.

Safety & tolerability with respect to opiate-like side effects

Despite their undisputable efficacy in acute pain, management using morphine and its derivatives elicit dose limiting unwanted side effects (nausea, constipation, sedation, respiratory depression, tolerance) while their long term use carries with it an elevated risk for increasing tolerance and addiction.

The following experiments were performed in animals to study these opiate side effects in case of use of DENKIs as a new pharmalogical class of painkillers.

Self-administration paradigm in mice

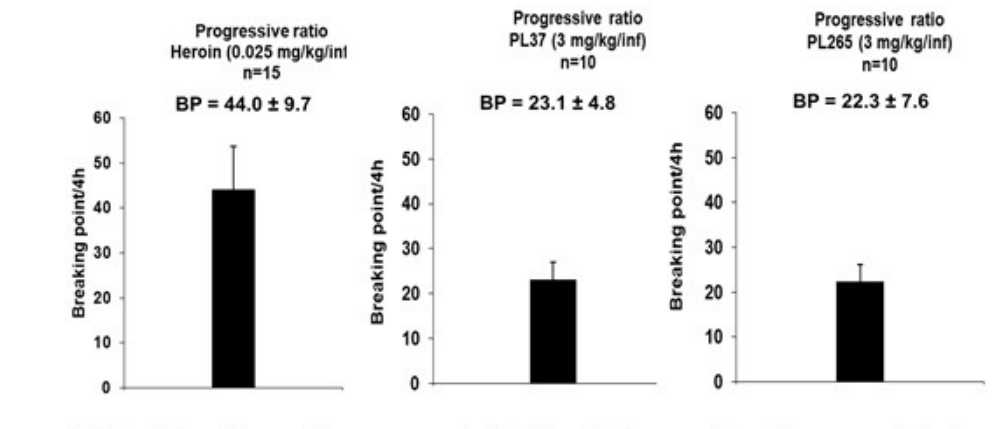
Progressive ratio Heroin (0.025 mg/kg/inf) n=15
BP = 44.0 ± 9.7
Progressive ratio PL37 (3 mg/kg/inf) n=10
BP = 23.1 ± 4.8
Progressive ratio PL265 (3 mg/kg/inf) n=10
BP = 22.3 ± 7.6
Breaking point/4h

The established breaking point value reveals that heroin has considerably more reinforcing effects than PL37 or PL265 in mice after intravenous administration, using a heroine dose more than 100 X lower than either PL37 or PL265.

Conditioned place preference in mice

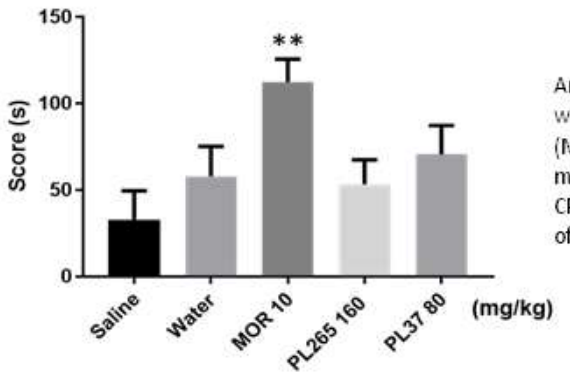
Score (s)
Saline
Water
MOR 10
PL265 160
PL37 80
(mg/kg)

Animals were conditioned during 4 days with saline, distilled water, morphine (MOR), PL37 and PL265. CPP was measured the 5th day.
CPP was conducted with the highest dose of PL265 and PL37.

After 5 days of conditioning, PL37 (80 mg/kg, ip) and PL265 (160 mg/kg, ip) induce no rewarding effects in mice, while morphine induces a significant effect (10 mg/kg ip).

No discriminative effects in rats

Rats were trained to discriminate between 0.3 mg/kg morphine and saline.

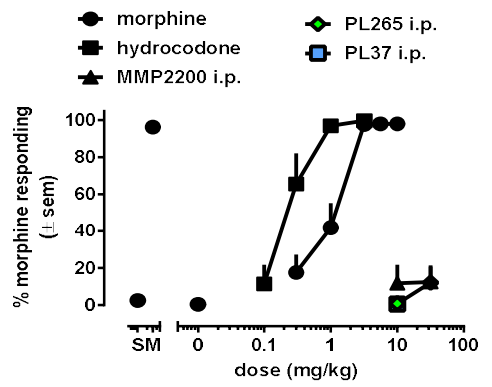
morphine
hydrocodone
MMP2200 i.p.
PL265 i.p.
PL37 i.p.
% morphine responding (± sem)
dose (mg/kg)

Morphine and hydrocodone produce dose-dependent increases in morphine-appropriate responding and decreases in rates of responding. Hydrocodone was slightly more potent than morphine. The opioid agonist MMP2200 with agonist activity at both mu- and delta-opioid receptors produces no drug lever selection in rats able to discriminate morphine from saline.

Rats were trained to discriminate between the delta-opioid receptor agonist SNC80 (3.2 mg/kg) and saline.

SNC80 produces dose-dependent increases in SNC80- appropriate responding. In addition, the delta-opioid receptor antagonist naltrindole attenuates the discriminative effects of SNC80 as demonstrated by rightward shifts in the SNC80 dose response curve. The mixed efficacy mu-opioid and delta-opioid receptor agonist MMP2200 produces no drug lever selection in rats able to discriminate SNC80 from saline.

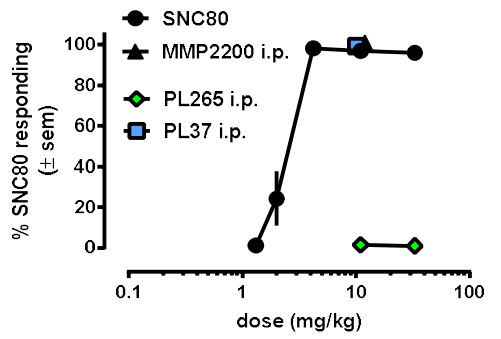
SNC80
MMP2200 i.p.
PL265 i.p.
PL37 i.p.
% SNC80 responding (± sem)
dose (mg/kg)

Neither PL37 nor PL265, at doses up to 10-32 mg/kg, elicit discriminating effects against morphine (µ opioid agonist) or SNC80 (δ opioid agonist).

No tolerance or cross-tolerance of PL37 with morphine in HPT model in mice

Repeated b.i.d. treatment with either saline, PL37 (80 mg/kg) (Left) or morphine (10 mg/kg) (Right) were performed by i.p. route for 4 consecutive days.

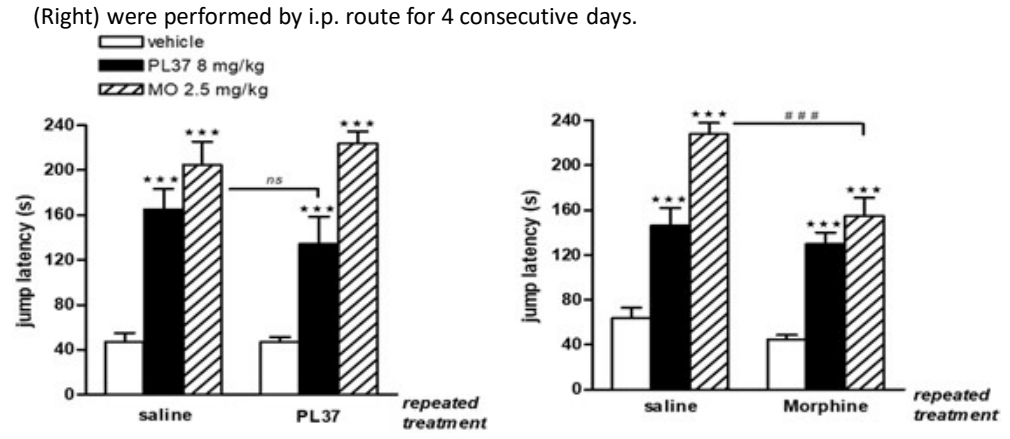
vehicle
PL37 8 mg/kg
MO 2.5 mg/kg
jump latency (s)
saline
PL37
Morphine
repeated treatment

No tolerance is observed with PL37 treatment (left) unlike morphine (right).
In case of morphine chronic treatment, no cross-tolerance of PL37 with morphine is observed.

No gastro-intestinal side effects in mice

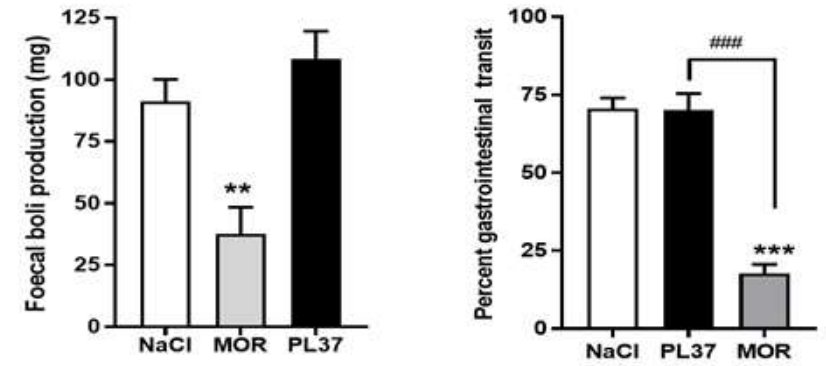
Foecal boli production (mg)
NaCl
MOR
PL37
Percent gastrointestinal transit

Unlike morphine (10 mg/kg s.c.) which produces a significant decrease in faeces production (left) and in transit time (right), PL37 (80 mg/kg s.c.) does not induce these G.I. side effects.

No constipation in humans during oral treatment

In a double-blind, placebo-controlled, cross-over Phase 1 study of PL37 *per os* on 28 healthy subjects at the **very high dose of 1000 mg q.i.d for 5 consecutive days** (the dose used in patients being 200 mg t.i.d.), no constipation was reported. On the contrary, 8 subjects (28%) presented diarrhea during the PL37 period vs 2 (7.1%) during the placebo period.[6]

In a double-blind, placebo-controlled study of PL37 at 200 mg t.i.d. *per os* for 28 days in patients with painful diabetic neuropathy, **no constipation was reported**, and diarrhea was reported in 5.4% of the patients on PL37 vs 3.7% of the patients on placebo.[6]

Discussion

PL265 is active *per os* in animal models of inflammatory and neuropathic pain. Adequately formulated, the DENKI PL37, administered intravenously is active in animal models of acute pain. The central antinociceptive effects of MO and endogenous PL37-protected ENKs lie within the same range, which is consistent with the similar affinity of MO and ENKs for MORs, primarily responsible of central modulation of pain.

Constipation, nausea, tolerance and addiction, some of the severe and unwanted side effects of MO were not observed with either DENKI. Interestingly the lack of cross-tolerance after chronic MO treatment is probably due to the internalization of MO-bound OR. PL37-protected ENKs favor OR-recycling. These results are in accordance with the recent study by Stoeber et al [8] which shows that whereas opioid peptides and alkaloids used to treat pain, such as morphine act on opioid receptors expressed at the cell plasma membrane, only alkaloids can activate intracellular receptors.

Altogether these new findings open the way to **the clinical use of PL37 and MO in rotation.**

Conclusions

Across a wide array of animal models of pain, DENKI induce antinociceptive responses similar to morphine. DENKI remain active after chronic administration of morphine, indicating that DENKI-induced analgesia persists in morphine-tolerant mice. Moreover, DENKI do not induce constipation in animals or humans. Finally, PL37 and PL265 are both shown here as devoid of any abuse liability even at high doses and after repeated administration and could be used as a new class of painkillers in all types of severe pain as an affordable solution to the opioid crisis.

Based on these remarkable pharmacological results, DENKIs are currently in clinical development as novel painkillers. PL265 is being developed for the oral treatment of neuropathic pain, inflammatory diseases of the bowel, osteoarthritis pain or topic treatment of ocular / Inflammatory pain whereas PL37 targets the treatment of post-surgical / traumatic / breakthrough cancer pain as a substitute to injectable opiates.

References:
1 Minett M.S., Pereira V., Sikandar S., Matsuyama A., Lolignier S., Kanellopoulos A.H., Mancini F., Iannetti G.D., Bogdanov Y.D., Santana-Varela S., et al., Nat Commun. 2015 Dec 4;6:8967. doi: 10.1038/ncomms9967
2 Roques B.P., Fournie-Zaluski M.C., and Wurm M.. Nature Reviews Drug Discovery, 2012. 11, 292-310.
3 Poras H., Bonnard E., Dangé E., Fournie-Zaluski M-C.,Roques B.P.. J. Med. Chem., 2014, *57*, 5748-5763.
4 Bonnard E., Poras H., Nadal X., Maldonado R., Fournié-Zaluski M.C., Roques B.P. Pharmacol. Res. Perspect., 2015, *3,* e00116, doi: 10.1002/prp2.116
5 Schreiter A., Core C,., Labuz D., Fournie-Zaluski MC., Roques B.P., Stein C., Machelska H., FASEB J., 2012, 26, 5161-5171
6 Pharmaleads unpublished results
7 Premiminary results will be presented to IASP 2018, Boston
8 Stoeber M., Jullié D., Lobingier B.T., Schiller P.W., Manglik A., von Zastrow M. Neuron, 2018, 98, 1-14